EMERALD-Y90: a Phase 2 study to evaluate transarterial radioembolization followed by durvalumab and bevacizumab for the treatment of unresectable hepatocellular carcinoma eligible for embolization

Riad Salem,[1] Anne M. Noonan,[2] Ben Spieler,[3] Sarah B. White,[4] Laura Kulik,[5] Dex Underwood,[6] Eric Heilbron,[6] Binh Nguyen,[6] Graham Wetherill,[7] Renuka Iyer[8]

[1]Northwestern University, Chicago, IL, USA; [2]The Ohio State University Comprehensive Cancer Center, Columbus, OH, USA; [3]University of Miami, Miami, FL, USA; [4]Medical College of Wisconsin, Milwaukee, WI, USA; [5]Northwestern Memorial Hospital, Chicago, IL, USA; [6]AstraZeneca, Gaithersburg, MD, USA; [7]AstraZeneca, Cambridge, UK; [8]Roswell Park Cancer Institute, Buffalo, NY, USA

Plain language summary

Why are we performing this research?

- For people with hepatocellular carcinoma (HCC) that cannot be removed with surgery, one of the preferred treatments is transarterial radioembolization (TARE). TARE delivers radiation beads to local blood vessels destroying the tumor
- Both durvalumab (a drug that helps the immune system fight cancer) and bevacizumab (a drug that starves the tumor by limiting its blood supply) have been shown to help people with HCC live longer when given as part of combination therapies
- The EMERALD-Y90 study is looking at the effects of treatment with TARE followed by durvalumab plus bevacizumab for people with HCC whose cancer cannot be removed surgically and has not spread outside of the liver

How are we performing this research?

- Participants will undergo a TARE procedure followed by durvalumab and bevacizumab treatment until the study concludes, the disease progresses, or there are unacceptable side effects
- The primary goal of this study is to measure the length of time after the TARE procedure that participants live without their cancer growing, spreading, or getting worse
- This study will also look at the types and frequency of side effects experienced by the participants

Who will participate in this study?

- Approximately 100 people with HCC whose cancer cannot be removed with surgery will be enrolled at one of approximately 20 sites across the US

Where can I access more information?

- This study (NCT06040099) is ongoing. We look forward to sharing the results once they are available
- More information about this study can be found at: https://clinicaltrials.gov/study/NCT06040099

This study was funded by AstraZeneca
Poster presented at the SIO 2024 Annual Scientific Meeting by Riad Salem

Background

- Liver cancer is the sixth most frequent cause of cancer-related deaths in the US,[1] with approximately 90% of primary liver cancer cases being HCC[2]
- Unresectable HCC that is eligible for embolization accounts for approximately 30% of HCC cases, and is commonly treated with locoregional therapy, such as transarterial chemoembolization (TACE) or TARE[3]
- Median progression-free survival (PFS) following TACE or TARE treatment is often less than one year for people with unresectable HCC amenable to embolization,[4,5] highlighting a need for additional treatment options

Rationale

- Locoregional therapies, such as TACE and TARE, are associated with response rates of approximately 50% per modified Response Evaluation Criteria in Solid Tumors (mRECIST)[6] and a median PFS of less than 1 year[7,8]
- Immune checkpoint inhibitors and VEGF-inhibitors have each been approved as treatments for unresectable HCC not amenable to locoregional therapy[9,10]
- Results from the Phase 3 EMERALD-1 study (NCT03778957) have shown that durvalumab (anti-programmed cell death ligand-1 antibody) in combination with TACE and bevacizumab (anti-VEGF antibody) demonstrated a statistically significant and clinically meaningful improvement in PFS, versus TACE alone, in patients with HCC eligible for embolization[11]
- A need still exists for evidence to support additional treatment options in settings where TARE is the preferred treatment modality
- The EMERALD-Y90 study (NCT06040099) will evaluate the efficacy and safety of TARE followed by durvalumab monotherapy (1 cycle) then durvalumab plus bevacizumab in participants with unresectable HCC amenable to embolization

EMERALD-Y90 study design: a Phase 2, single-arm, US-only, multicenter, practice-informing study to assess the efficacy and safety of TARE followed by durvalumab monotherapy followed by durvalumab + bevacizumab in unresectable HCC amenable to embolization

Study begins | Day 1 | Day 15 + 3 after TARE | Day 14 + 3 after durvalumab

Screening and enrollment (Day 0 + 28)

~100 participants with unresectable HCC amenable to embolization

▶ TARE + post-TARE single-photon emission computed tomography (SPECT-CT) within 24hrs ▶ (14 + 3 days) Durvalumab 1500 mg (single dose) ▶ (14 + 3 days) Durvalumab 1120 mg + bevacizumab 15 mg/kg Q3W ▶ Progression, unacceptable toxicity, or study end

Pre-TARE dosimerty

No contraindications for durvalumab

No contraindications for bevacizumab

A pre-specified safety analysis will occur approximately 2 months after the first 30 patients initiate durvalumab treatment

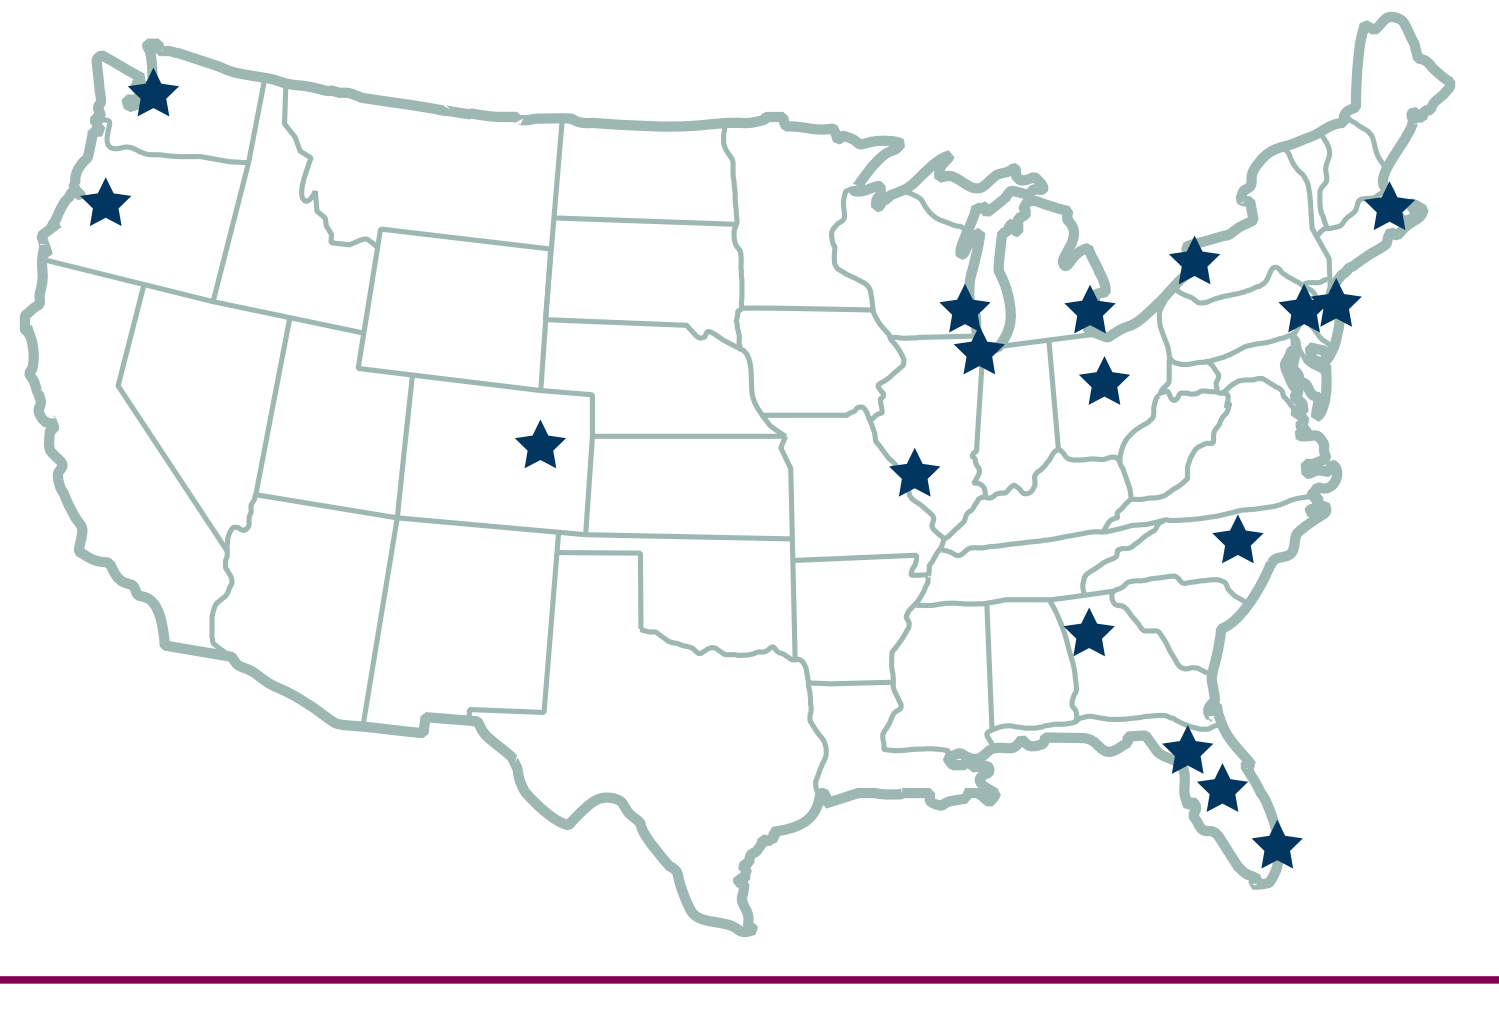

Recruiting in the US only

Approximately 20 study sites

Currently enrolling patients

Expected study end: June 2027

Key inclusion criteria

- Age ⩾18 years old
- Confirmed unresectable HCC (by imaging or histopathological findings from biopsy specimen) that is amenable to treatment with TARE, and the entire tumor volume can be treated in one session
- No evidence of extrahepatic spread
- One or more measurable lesions, unilobar disease for participants with Vp1/Vp2 portal vein invasion and eligible for Y90 TARE
- Child-Pugh class A liver function
- Eastern Cooperative Oncology Group performance status 0–1 at enrollment
- Adequate organ and marrow function

Key exclusion criteria

- Disease amenable to curative surgery, transplantation, or curative ablation
- Coinfection with HBV and HDV
- Prior systemic therapy for HCC
- HCC requiring more than one TARE session
- Receipt of prior locoregional therapy (previous TACE, TARE, or stereotactic body radiation therapy associated with the curative setting more than 6 months prior to study is permitted, and radiofrequency ablation is permitted if the target lesion was not treated or had subsequently progressed)
- Receipt of more than one prior embolization procedure (TACE or TARE)
- Angiogram showing less than 30% future liver remnant volume
- Major portal vein invasion (Vp3/Vp4)

Study endpoints

1°

- **PFS:** time from TARE until disease progression, assessed by the investigator per mRECIST, or death due to any cause

2°

- **Safety and tolerability:** incidence, severity, nature, seriousness, and causality of adverse events, vital signs, clinical laboratory assessments, and electrocardiograms from day of TARE, were among the safety outcomes assessed
- **Objective response rate:** proportion of participants with a confirmed complete or partial response, assessed by the investigator per mRECIST
- **Overall survival:** time from TARE until death due to any cause
- **Duration of response:** time from first documented response until date of progression, assessed by the investigator per mRECIST, or death due to any cause

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please click on the link below.

Poster

https://bit.ly/48CZ7nH

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the authors of this poster.

Abbreviations

HBV, hepatitis B virus; HCC, hepatocellular carcinoma; HDV, hepatitis D virus; mRECIST, modified Response Evaluation Criteria in Solid Tumors; PFS, progression-free survival; Q3W, every 3 weeks; SPECT-CT, single-photon emission computed tomography; TACE, transarterial chemoembolization; TARE, transarterial radioembolization; VEGF, vascular endothelial growth factor; Y90, Yttrium-90.

Acknowledgments

This study is sponsored by AstraZeneca. AstraZeneca would like to acknowledge additional support from Philip Sinclair (Boston Scientific), Aaron Bartoo (Boston Scientific), Gordon Cohen (AZ), and Kerry Parsons (AZ). Medical writing support, under the direction of the authors, was provided by Andrea Hough, PhD, CMC Connect, a division of IPG Health Medical Communications, funded by AstraZeneca, in accordance with Good Publication Practice (GPP 2022) guidelines.

Disclosures

RS and LK report being a consultant for AstraZeneca. SBW reports being a steering committee member for AstraZeneca. DU, EH, and BN are employees and hold stock in AstraZeneca. GW is a contractor for AstraZeneca. AMN, BS, and RI report no conflicts of interest related to AstraZeneca. Full author disclosures are available with the published abstract.

References

1. American Cancer Society. Cancer Statistics Center. https://cancerstatisticscenter.cancer.org/. Accessed December 6, 2023.
2. European Association for the Study of the Liver. *J Hepatol*. 2018;69:182–236.
3. Prince D, et al. *Ther Adv Med Oncol* 2020;12:1–17.
4. Teyateeti A, et al. *J Hepatocell Carcinoma* 2020;7:117–131.
5. Li S, et al. *J Cancer* 2019;10:5007–5014.
6. Moreno-Luna LE, et al. *Cardiovasc Intervent Radiol* 2013; 36:714–723.
7. Pitton MB, et al. *Cardiovasc Intervent Radiol* 2015;38:352–360.
8. Akinwande O, et al. *Anticancer Res* 2016;36:239–246.
9. AstraZeneca. Imfinzi® (durvalumab): highlights of prescribing information. 2023. https://www.azpicentral.com/pi.html?product=imfinzi. Accessed December 18, 2023.
10. Genentech Inc. Avastin® (bevacizumab): Highlights of prescribing information. 2022. https://www.gene.com/download/pdf/avastin_prescribing.pdf. Accessed December 18, 2023.
11. AstraZeneca. Press Release. Published November 9, 2023. https://www.astrazeneca.com/media-centre/press-releases/2023/imfinzi-combination-improves-pfs-in-liver-cancer.html. Accessed December 6, 2023.

Supported by: AstraZeneca

Poster presented at: Society of Interventional Oncology Annual Scientific Meeting, Long Beach, CA, USA, January 25–29, 2024. Corresponding author email address: Riad Salem (rsalem1@nm.org)